L15

Effectiveness of Sebetralstat for the On-demand Treatment of Laryngeal Hereditary Angioedema Attacks: Interim Analysis from KONFIDENT-S

Jonathan A. Bernstein,[1] Emel Aygören-Pürsün,[2] Vesna Grivcheva-Panovska,[3] Inmaculada Martinez-Saguer,[4] Danny M. Cohn,[5] William R. Lumry,[6] Marc A. Riedl,[7] Andrea Zanichelli,[8,9] James Hao,[10] Michael D. Smith,[10] Christopher M. Yea,[10] Paul K. Audhya,[10] Henriette Farkas[11]

[1]University of Cincinnati College of Medicine and Bernstein Clinical Research Center, Cincinnati, OH, USA; [2]University Hospital Frankfurt, Goethe University Frankfurt, Frankfurt, Germany; [3]University Clinic of Dermatology, School of Medicine, University Saints Cyril and Methodus, Skopje, North Macedonia; [4]HZRM Haemophilia Center Rhein Main, Frankfurt, Germany; [5]Amsterdam University Medical Center, University of Amsterdam, Amsterdam, Netherlands; [6]AARA Research Center, Dallas, TX, USA; [7]University of California, San Diego, La Jolla, CA, USA; [8]IRCCS Policlinico San Donato Milanese, Milan, Italy; [9]University of Milan, Milan, Italy; [10]KalVista Pharmaceuticals, Salisbury, United Kingdom, and Cambridge, MA, USA; [11]Hungarian Angioedema Center of Reference and Excellence, Semmelweis University, Budapest, Hungary

Background

- Although they represent roughly 1% of all swelling attacks, attacks involving the larynx occur in >50% of people living with hereditary angioedema (HAE) type 1 or type 2 due to C1-inhibitor deficiency (HAE-C1INH) at least once in their lifetime[1-3]
- These attacks may be fatal without prompt administration of an appropriate on-demand treatment[1-3]
 - In a previously reported case series, laryngeal attacks were rarely noted to be lethal in less than 10 minutes; death occurred within 20 minutes in 4 of 32 fatal laryngeal attacks (11%)[4]
- Currently approved therapies for on-demand treatment of HAE-C1INH attacks must be administered parenterally; the treatment burden of parenteral administration is associated with delays and/or withholding of treatment[5-7]
- In a recent survey of people living with HAE in the United States, the mean time to treatment of attacks involving the larynx and/or tongue was 2.5 hours; 25% of respondents reported waiting 5 hours or more[8]
- Sebetralstat, an investigational oral plasma kallikrein inhibitor, is being evaluated as on-demand treatment for HAE attacks in the ongoing, 2-year, multicenter, open-label extension (OLE) study KONFIDENT-S (NCT05505916, EudraCT: 2021-001176-42) that was designed to align with guidelines recommending early treatment[9]

Objective

- The objectives of this interim analysis of KONFIDENT-S are to assess the tolerability, safety, and effectiveness of sebetralstat as an on-demand treatment for HAE attacks involving the larynx

Methods

Study Design

- Eligible participants were adults (≥18 years of age) and adolescents (≥12 years of age) with HAE-C1INH and ≥2 documented attacks within 3 months or who completed the phase 3 KONFIDENT trial
 - Participants receiving long-term prophylaxis (LTP) were required to be on a stable dose and regimen for ≥3 months immediately before and during the study
- Consistent with the EAACI/WAO treatment guideline recommendations for HAE,[9] participants were instructed to self-administer treatment (sebetralstat 600 mg) as early as possible after attack onset
 - Participants were instructed to treat immediately with conventional on-demand treatment if laryngeal attack symptoms worsened after the initial administration of sebetralstat
- Effectiveness endpoints were as follows:
 - Time to beginning of symptom relief (Patient Global Impression of Change rating of at least “A Little Better” for ≥2 consecutive time points) within 12 hours
 - Time to reduction in attack severity (≥1 level decrease on the Patient Global Impression of Severity [PGI-S] for ≥2 consecutive time points) within 12 hours
 - Time to complete attack resolution (PGI-S rating of “None”) within 24 hours
- Conventional treatment administration was censored to the end of the analysis window

Results

Participant Characteristics

- From October 21, 2022, to September 14, 2024 (data cutoff), 134 participants treated 1706 attacks with sebetralstat 600 mg (**Table 1**)
 - 32 of these 1706 attacks (1.9%) involved the larynx (**Table 2**)
 - The median time from attack onset to treatment was 11.5 minutes (IQR, 1.0 to 34.0) for attacks involving the larynx

Table 1. Participant Demographics

	All participants N=134	Participants experiencing attacks involving the larynx n=16
Age, median (IQR), years	35.0 (21.0 to 49.0)	44.5 (29.5 to 59.5)
Age group, n (%)		
≥12 to <18 years	23 (17.2)	2 (12.5)
≥18 to <65 years	107 (79.9)	13 (81.3)
≥65 years	4 (3.0)	1 (6.3)
Sex, female, n (%)	87 (64.9)	10 (62.5)
Race, n (%)		
White	99 (73.9)	13 (81.3)
Asian	17 (12.7)	2 (12.5)
Other or multiple	11 (8.2)	–
Not reported	7 (5.2)	1 (6.3)
BMI, median (IQR), kg/m²	25.4 (22.3 to 30.6)	29.6 (26.4 to 32.2)
HAE-C1INH type, n (%)		
Type 1	125 (93.3)	16 (100)
Type 2	9 (6.7)	–
Current treatment regimen, n (%)		
On-demand only	99 (73.9)	9 (56.3)
LTP + on-demand	35 (26.1)[a]	7 (43.8)

BMI, body mass index; HAE-C1INH, hereditary angioedema type 1 or type 2 due to C1-inhibitor deficiency; IQR, interquartile range; LTP, long-term prophylaxis.
[a]Of the 35 participants using LTP, 29 (21.6%) used kallikrein inhibitors (berotralstat or lanadelumab) and 6 (4.5%) used C1INH replacement.

Table 2. Baseline Attack Characteristics

	All attacks treated with sebetralstat N=1706	Attacks involving the larynx treated with sebetralstat n=32
Baseline PGI-S category,[a] n (%)		
Mild	589 (34.5)	7 (21.9)
Moderate	685 (40.2)	15 (46.9)
Severe/very severe	330 (19.3)	9 (28.1)
Time from attack onset to treatment administration, median (IQR), minutes	10.0 (1.0 to 68.5)	11.5 (1.0 to 34.0)

IQR, interquartile range; PGI-S, Patient Global Impression of Severity.
[a]Missing: n (%) = 73 (4.3) for all attacks; None: n (%) = 29 (1.7) for all attacks, 1 (3.1) for attacks involving the larynx treated with sebetralstat.

Tolerability and Safety

- No participants reported difficulty swallowing sebetralstat during an attack involving the larynx
- 7 participants treating attacks involving the larynx with sebetralstat experienced treatment-emergent adverse events (TEAEs) (**Table 3**)
- 2 TEAEs were considered treatment-related: 1 instance each of grade 2 nausea and grade 2 vomiting in the same participant, who experienced an attack involving the larynx and abdomen
- 2 TEAEs of "laryngeal HAE attack" in 1 adolescent resulted in hospitalization (precautionary) and were not considered related to treatment

Table 3. Safety

TEAE, n (%)	Participants treating attacks involving the larynx n=16
Any TEAE	7 (43.8)
Treatment-related	**1 (6.3)**
Serious TEAE	2 (12.5)[a]
Treatment-related	**0**
Severe TEAE	3 (18.8)
Treatment-related	**0**
Any TEAE leading to permanent discontinuation	1 (6.3)
Any TEAE leading to death	0
Treatment-related TEAEs	
Gastrointestinal disorders	
Nausea	1 (6.3)[b]
Vomiting	1 (6.3)[b]

E, number of events; TEAE, treatment-emergent adverse event.
[a]Serious TEAEs were 1 event of grade 3 viral meningitis occurring in 1 participant and 2 events of laryngeal HAE attack occurring in 1 participant.
[b]Grade 2 nausea and vomiting occurred in the same participant.

Effectiveness

Figure 1. Effectiveness Endpoints

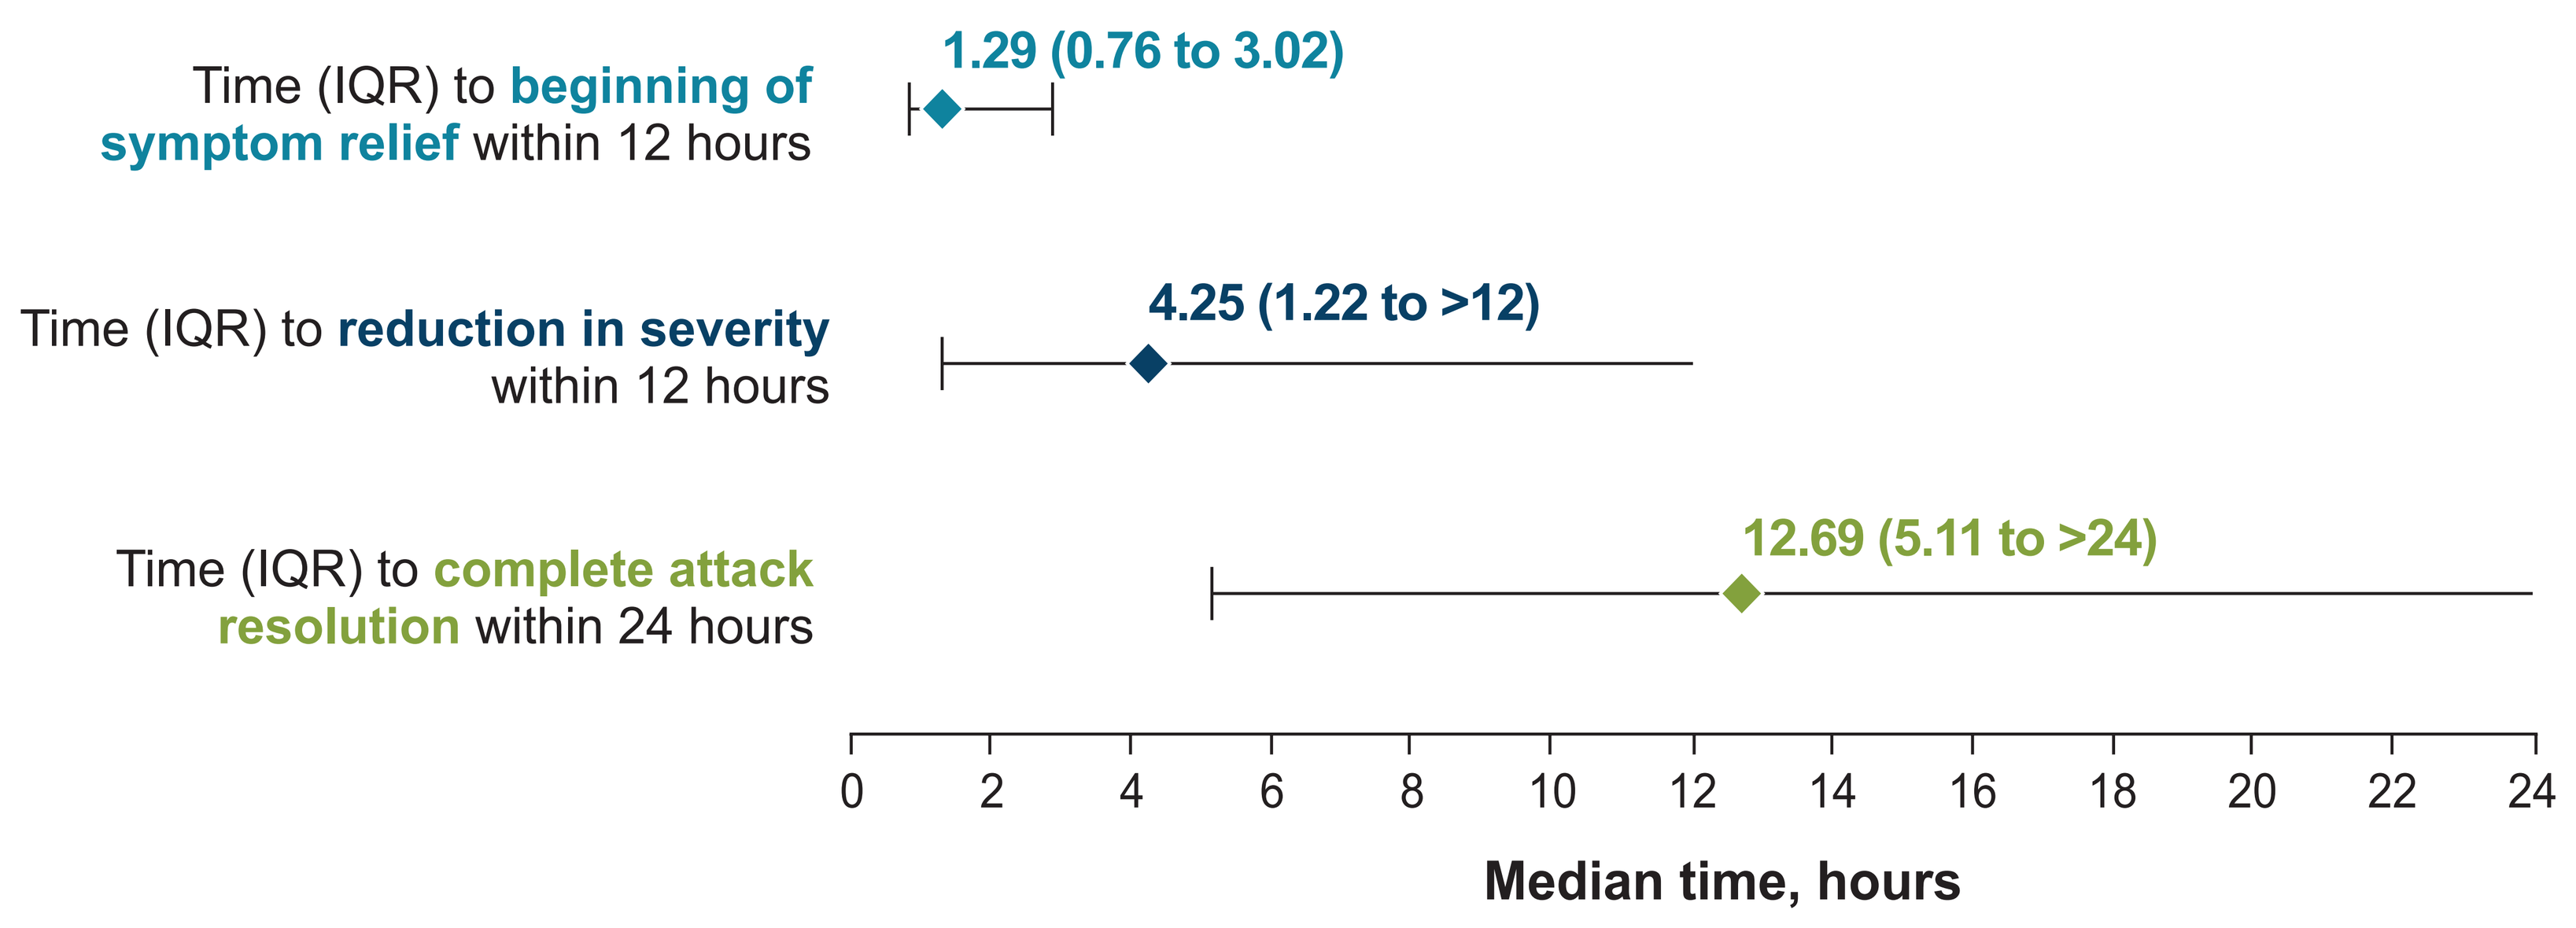

Time (IQR) to beginning of symptom relief within 12 hours
1.29 (0.76 to 3.02)
Time (IQR) to reduction in severity within 12 hours
4.25 (1.22 to >12)
Time (IQR) to complete attack resolution within 24 hours
12.69 (5.11 to >24)
0 2 4 6 8 10 12 14 16 18 20 22 24
Median time, hours

IQR, interquartile range.
Error bars represent IQR.

- 4 of the 32 attacks involving the larynx (12.5%) were treated with a second administration of sebetralstat within 12 hours and 3 (9.4%) were treated with conventional treatment within 12 hours; the time of conventional treatment administration was not recorded for 1 attack
 - Conventional treatment was administered after a single administration of sebetralstat in 2 attacks; 1 participant reported administering conventional on-demand treatment 1 minute after sebetralstat administration (PGI-S at the time of treatment was rated as very severe)
 - For 1 attack, conventional treatment was administered after 2 administrations of sebetralstat, 11.7 hours after the first administration of sebetralstat
- Most of the attacks (96%) affecting the larynx which achieved beginning of symptom relief did so without an additional administration of sebetralstat (**Figure 2**)

Figure 2. Proportion of Attacks Achieving Beginning of Symptom Relief Prior to or Without an Additional Administration of Sebetralstat

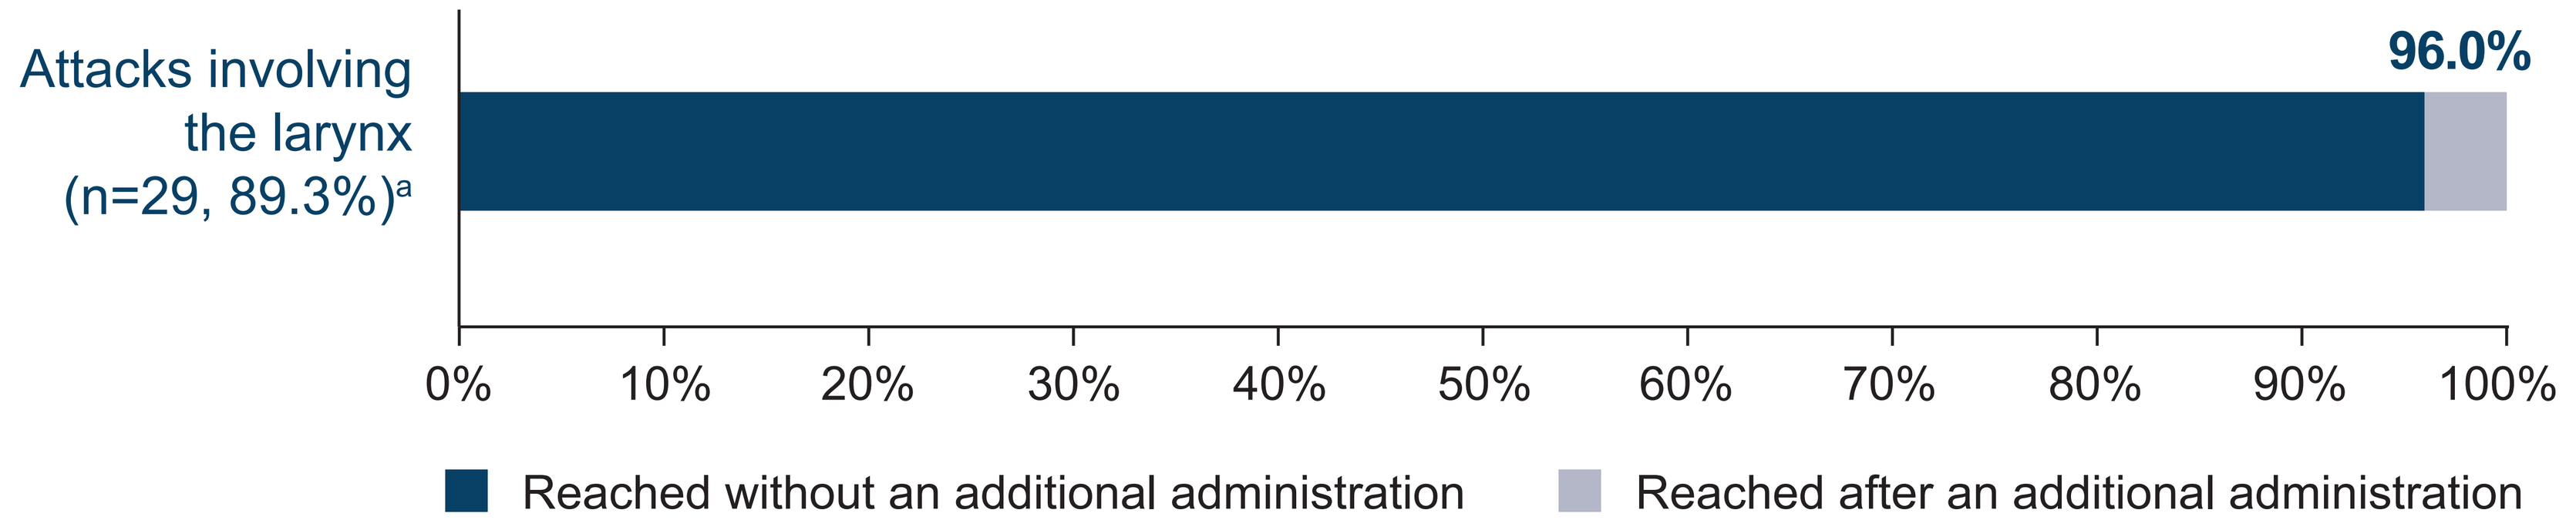

Attacks involving the larynx (n=29, 89.3%)[a]
96.0%
0% 10% 20% 30% 40% 50% 60% 70% 80% 90% 100%
Reached without an additional administration
Reached after an additional administration

[a]Proportion of attacks reaching the endpoint within 12 hours.

Conclusions

- **Sebetralstat was well-tolerated for attacks involving the larynx**
 - **No participants reported trouble swallowing sebetralstat**
- **Oral sebetralstat enabled early treatment (median 11.5 minutes) and resulted in early symptom relief (median 1.29 hours)**
- **A second administration of sebetralstat was used in 12.5% of attacks; conventional treatment was used in 9.4%**
- **Most of the attacks (96%) involving the larynx that achieved beginning of symptom relief did so without an additional administration of sebetralstat**

References

1. Bork K et al. *Allergy Asthma Clin Immunol*. 2021;17:40.
2. Bork K et al. *J Emer Med*. 2015;50:567-580.
3. Zuraw BL. *N Engl J Med*. 2008;359:1027-1036
4. Bork K et al. *J Allergy Clin Immunol*. 2012;130:692-697
5. Gower RG, Wilber M. *Allergy Asthma Clin Immunol*. 2021;17:100.
6. Maurer M et al. *PLoS One*. 2013;8:e53773.
7. Mendivil J et al. *Orphanet J Rare Dis*. 2021;16:94.
8. Christiansen S et al. *Ann Allergy Asthma Immunol*. 2024 (in press).
9. Maurer M et al. *Allergy*. 2022;77:1061-1990.

Acknowledgments

Medical writing assistance was provided under the direction of the authors by Richard W. Davis IV, PhD, of ApotheCom, San Francisco, CA, USA, and was funded by KalVista Pharmaceuticals.

Contact Information

Contact the author at bernstja@ucmail.uc.edu for questions or comments.

Please scan the QR code to view this poster after the presentation.

Presented at the American Academy of Allergy, Asthma, and Immunology/World Allergy Organization Joint Conference; February 28-March 3, 2025; San Diego, California

Copyright © 2025 KalVista Pharmaceuticals, Inc. All rights reserved.